Efficacy and tolerability of ADX10059, a negative allosteric modulator of mGluR5, on gastro-esophageal reflux: pH-impedance study in healthy subjects

Frank Zerbib, CHU St André, Bordeaux, France, Charlotte Keywood, Addex Pharma S.A., Switzerland

Background and Rationale

- Animal studies have shown that inhibition of mGluR5 reduces TLESRs and increases LES tone. ADX10059 is a negative allosteric modulator (NAM) of mGluR5. It binds to the receptor trans-membrane domain distant from the orthosteric glutamate binding site to inhibit receptor activity in response to the natural ligand.
- A preliminary study with ADX10059 in GERD patients[1] demonstrated a reduction in 24 hour oesophageal acid exposure and clinical symptoms on a single day of dosing, but with suboptimal tolerability, due to rapid absorption of the compound after oral dosing.

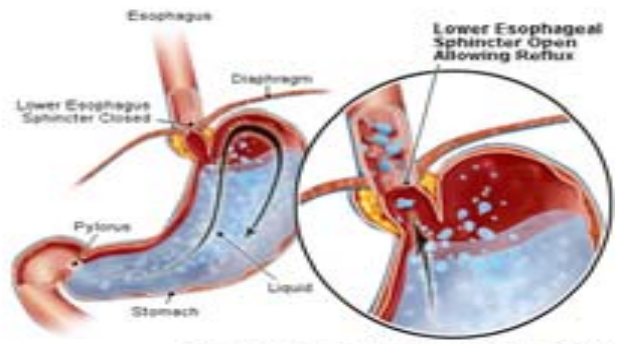
Esophagus
Lower Esophageal Sphincter Open Allowing Reflux
Diaphragm
Lower Esophagus Sphincter Closed
Pylorus
Liquid
Stomach

Gastroesophageal Reflux

- The aim of this study was to evaluate the pharmacokinetics, tolerability and pharmacodynamics of a modified release (MR) formulation of ADX10059.

Study Design

- The study was a single-center, randomized, double-blind placebo-controlled, multiple ascending dose study of the safety, pharmacokinetics and pharmacodynamics of three dose levels of ADX10059 in 24 healthy male subjects aged 18 to 55 years.
- Subjects took a single dose on Day 1 and 6 and twice daily doses on Days 2 to 5.
- Three dose groups of 50, 125 and 250 mg; n = 8 with 6 active and 2 placebo subjects per group
- Oesophageal pH impedance monitoring was performed pre dose on Day -1 and on dosing Day 6. Monitoring was performed for 5 hours. For one hour fasting and then for 4 hours (2 hours supine and 2 hours upright) following a standardized refluxogenic meal. Traces were analyzed by central blinded observation.

1 Keywood C, Wakefield M, Tack J, Gut 2009, in press

Study Evaluations

Pharmacokinetics of repeated dose administration of the modified release formulation of ADX10059.

Safety and tolerability of 6 days administration of the three doses of ADX10059

Pharmacodynamics: pH impedance measured reflux events in the Total, Post Prandial, Upright and Supine periods.
The primary efficacy variables were evaluated for the Total Period and comprised:
• Number of Reflux Episodes
• Number of Acidic Reflux Episodes
• Number of Weakly Acidic Reflux Episodes
• Total Acid Exposure %
• Total Bolus Exposure %

The change from baseline (Day 6 – Day -1) was compared for the treatment groups. Treatment effect was determined by the Kruskall Wallis test and comparison to placebo by the Wilcoxon rank sum test

Results

Pharmacokinetics
• Compared to the immediate release capsule, the MR formulation reduced Cmax by approximately 40% , increased Tmax from 1 hour to 4 hours, whilst maintaining the same AUC. Twice daily dosing resulted in satisfactory 24 hour plasma exposure at each dose level.

Pharmacodynamics – pH impedance monitoring
• There was a tendency for reflux episodes to increase on Day 6 compared to Day -1. There was a significant overall treatment effect for Total Acid Exposure % (p = 0.048) and total number of Weakly Acidic Reflux Episodes (p =0.041)

• Significant differences compared to placebo were seen for all primary efficacy variables except Number of Acid Reflux Episodes, for the 125mg bid dose. The median change from baseline for Acid Reflux Episodes (N) was placebo 3, 50mg 2, 125mg 1 and 250mg 3.5.

• Non significant trends in reducing reflux parameters were seen for the 250mg bid dose which was not more effective than 125 mg bid. The 50mg dose was not significantly superior to placebo.

The primary efficacy variables are summarized Figures 1 to 4.

Figure 1: Total reflux episodes (N)
Median change from baseline

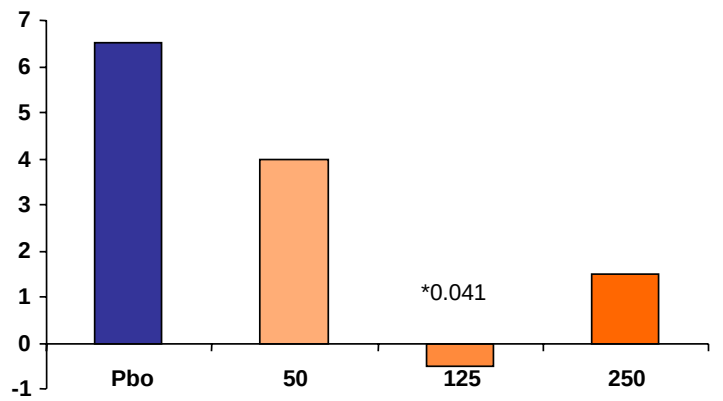
7
6
5
4
3
2
1
0
-1
*0.041
Pbo
50
125
250

Figure 2: Weakly acidic reflux episodes (N)
Median change from baseline

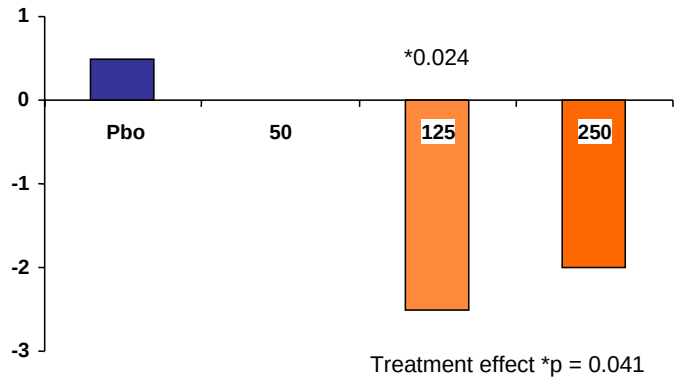
1
0
-1
-2
-3
*0.024
Pbo
50
125
250
Treatment effect *p = 0.041

Safety and tolerability

- No safety monitoring abnormalities (ECG, bloods, HR, BP) were observed
- The MR formulation improved the tolerability of ADX10059
- Adverse event frequency was 50 mg : n=3 ; 125 mg : n=8 ; 250 mg : n=9. All were mild or moderate. The most common were insomnia (n=4), constipation (n=2) and flatulence (n=2).

Figure 3: Total acid exposure %
Median change from baseline

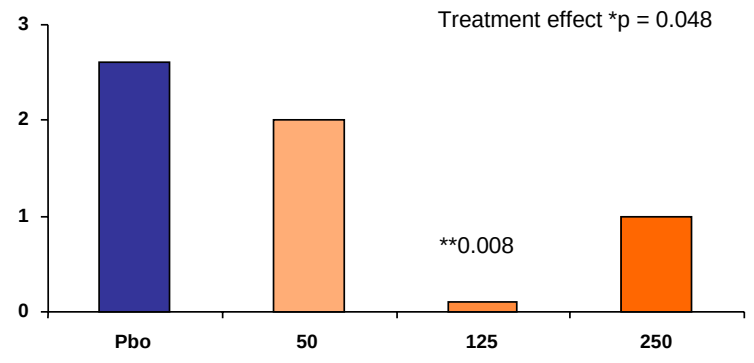
Treatment effect *p = 0.048
3
2
1
0
**0.008
Pbo
50
125
250

Total bolus exposure %
Median change from baseline

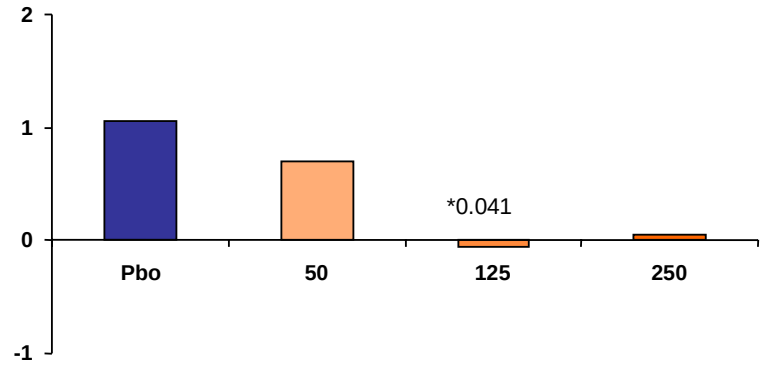
2
1
0
-1
*0.041
Pbo
50
125
250

Conclusions

- ADX10059 decreased gastro-oesophageal reflux episodes in healthy subjects
- The results of this study in healthy subjects support the findings of the previous study in GERD patients.
- The modified release formulation demonstrated pharmacokinetics suitable for twice daily administration
- The tolerability of the MR formulation is suitable for longer term treatment to evaluate clinical symptom control in GERD patients..